AZD0466, a nanomedicine of a potent dual Bcl-2/Bcl-xL inhibitor, exhibits anti-tumor activity in a range of haematological and solid tumor models

Srividya B. Balachander[1], Areya Tabatabai[1], Shenghua Wen[1], Steven Criscione[1], Francis D. Gibbons[2], Giulia Fabbri[3], David Jenkins[1], Courtney Anderson[1], Jeffery Tyner[4], Steven Kurtz[4], Guangnong 'Sunny' Zhang[2], Justin Cidado[1], Alwin Schuller[1], Lorraine Graham[5], Marianne Ashford[6], and Barry Davies[7]

[1]Bioscience, [2]DMPK, [3]Translational Medicine, Early Oncology, AstraZeneca, Boston, USA, [4]Knight Cancer institute, OHSU, [5]CMC Projects, [6]Advanced Drug Delivery, Biopharmaceuticals R&D, AstraZeneca, Macclesfield, UK, [7]Projects, Oncology R&D, Research & Early Development, AstraZeneca, Cambridge, UK

P - 56

Abstract

The induction of apoptosis in tumor cells represents a promising approach to the treatment of cancer. In tumor cells, the B cell lymphoma 2 (Bcl-2) protein family promotes cell survival through upregulation of anti-apoptotic Bcl-2 proteins, such as Bcl-2, Bcl-xL, Mcl-1 and Bfl-1. Clinical activity of the Bcl-2 inhibitor venetoclax has validated the approach of targeting this class of molecules, but additional value remains in jointly targeting Bcl-2 with other family members. AZD0466 is a novel drug-dendrimer conjugate, where the active moiety, AZD4320, is chemically conjugated to Starpharma's clinically validated DEP® dendrimer platform, a 5-generation PEGylated poly-lysine dendrimer via a hydrolytically labile linker. AZD4320 is a dual Bcl-2/Bcl-xL inhibitor that is equipotent to venetoclax and 3-fold more potent than navitoclax. AZD0466 has been optimized to maintain efficacy whilst mitigating anticipated on-target toxicities of AZD4320[1].

Introduction

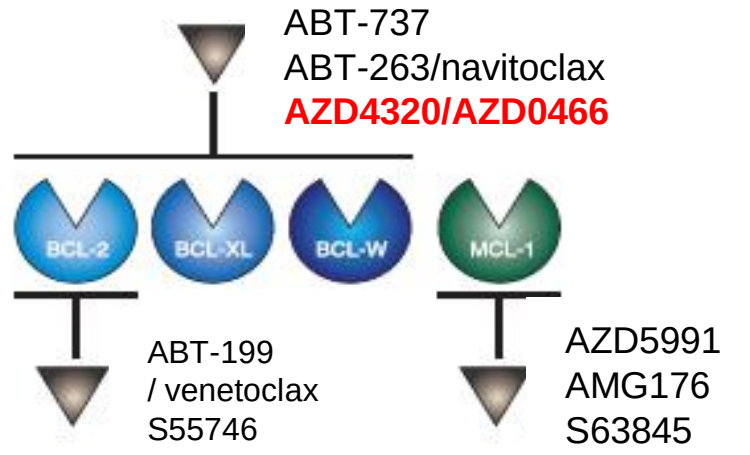
ABT-737
ABT-263/navitoclax
AZD4320/AZD0466
BCL-2
BCL-XL
BCL-W
MCL-1
ABT-199
/ venetoclax
S55746
AZD5991
AMG176
S63845

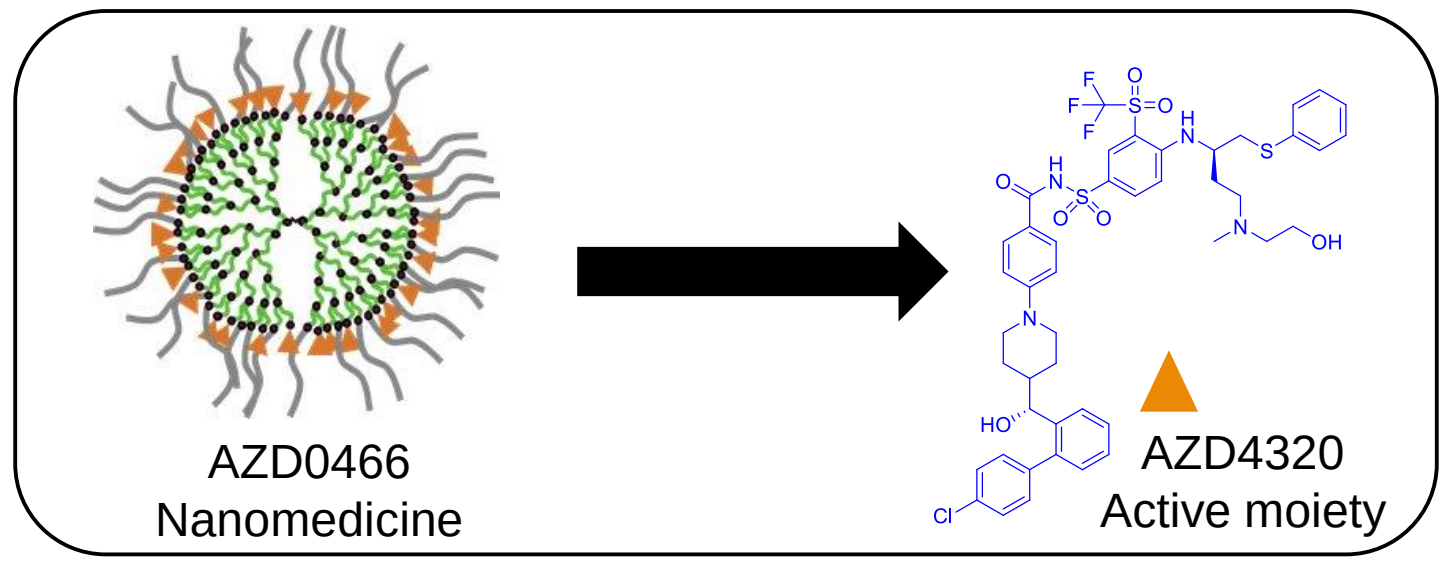
AZD0466
Nanomedicine
AZD4320
Active moiety

- AZD0466: a dendrimer-conjugate of AZD4320, small molecule inhibitor of Bcl-2 and Bcl-xL
- Enhanced solubility compared to AZD4320
- Once weekly dosing schedule with PK properties optimized to maximize therapeutic index

Results

AZD0466 active moiety potently reduces viability of heme cancer cell lines, SCLC cell lines and primary leukemic cells

A: Sanger Panel

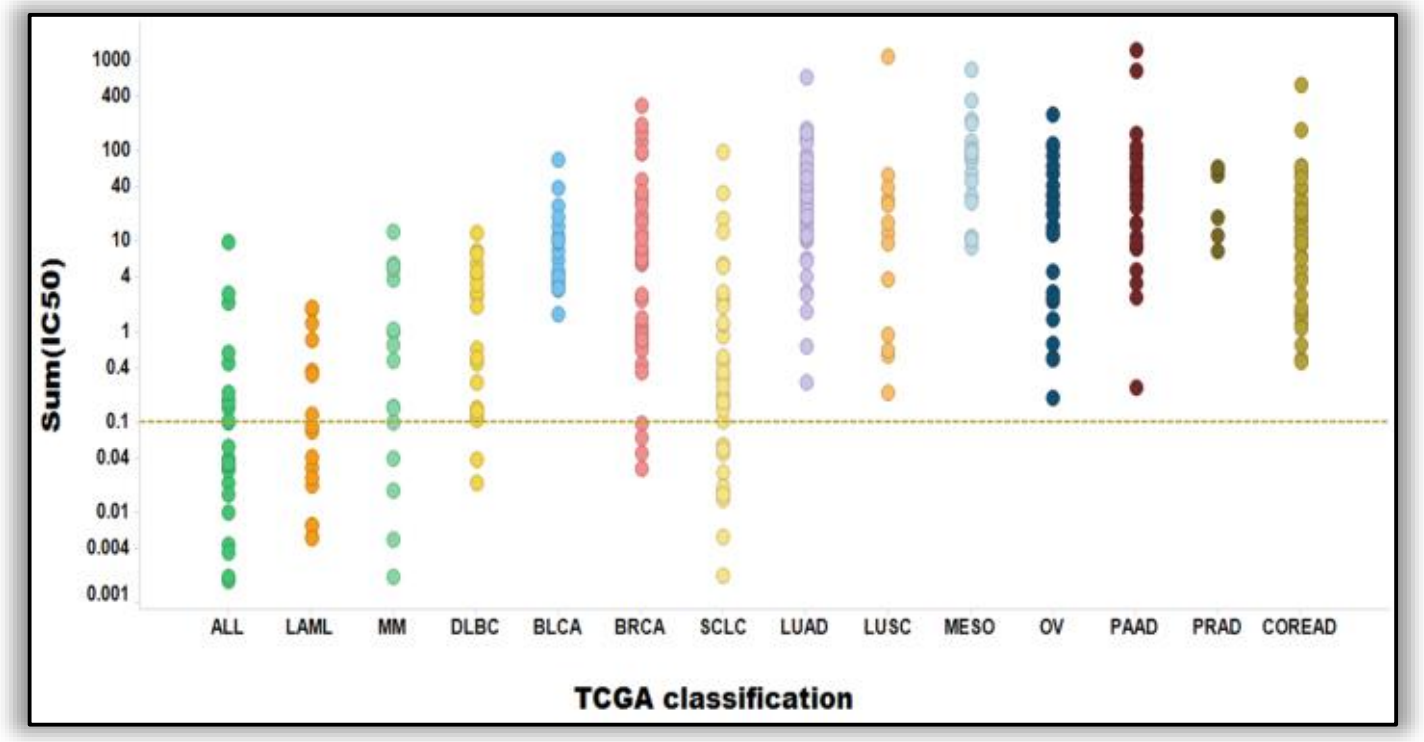
Sum(IC50)
1000
400
100
40
10
4
1
0.4
0.1
0.04
0.01
0.004
0.001
ALL
LAML
MM
DLBC
BLCA
BRCA
SCLC
LUAD
LUSC
MESO
OV
PAAD
PRAD
COREAD
TCGA classification

B: Beat AML Screen

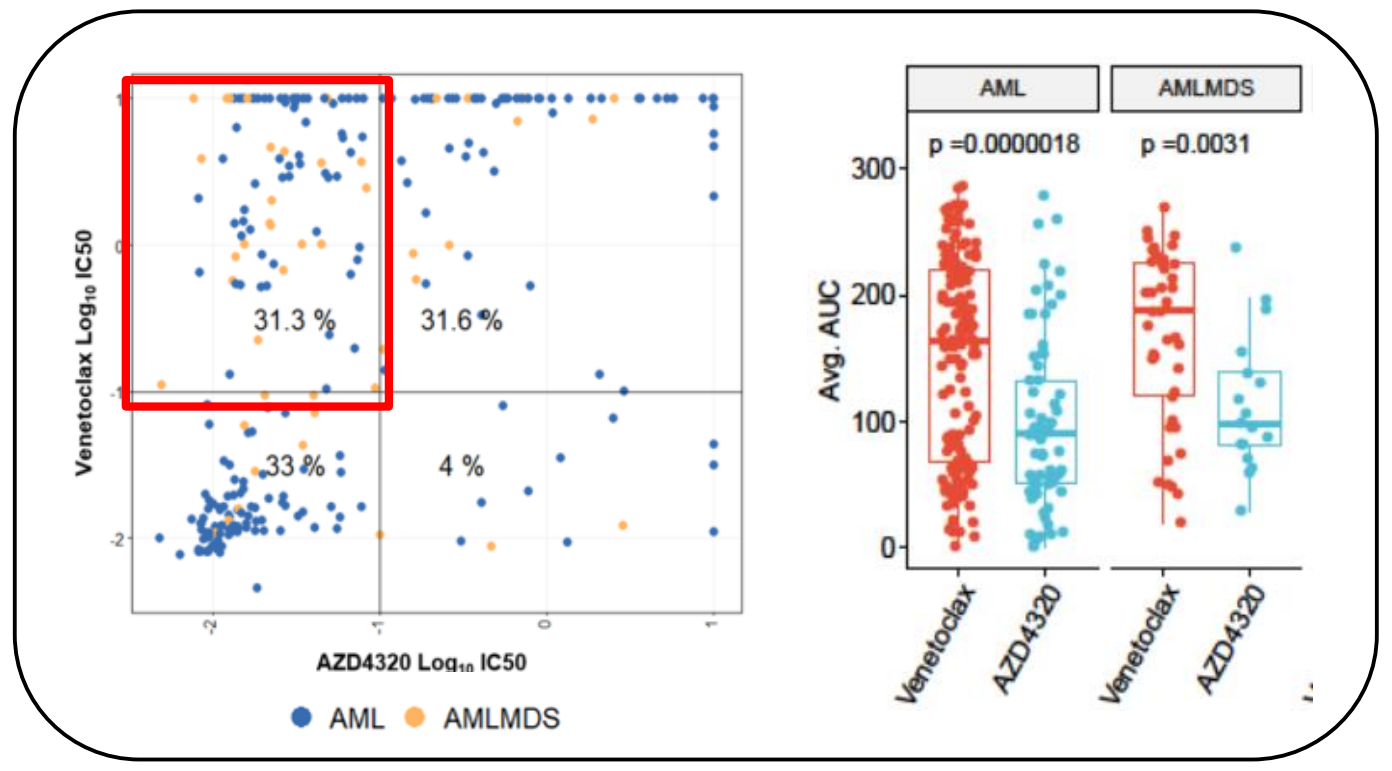
Venetoclax Log10 IC50
AZD4320 Log10 IC50
31.3 %
31.6 %
33 %
4 %
AML
AMLMDS
AML
AMLMDS
p =0.0000018
p =0.0031
Avg. AUC
300
200
100
0
Venetoclax
AZD4320
Venetoclax
AZD4320

(A) Subset of cancer cell lines sensitive to single agent AZD4320 (active moiety of AZD0466) in Sanger panel. (B) Broader activity of AZD4320 in primary AML patient samples compared to venetoclax (red box).

AZD0466 has potent antitumor activity in preclinical hematological models

A: RS4;11 ALL xenograft

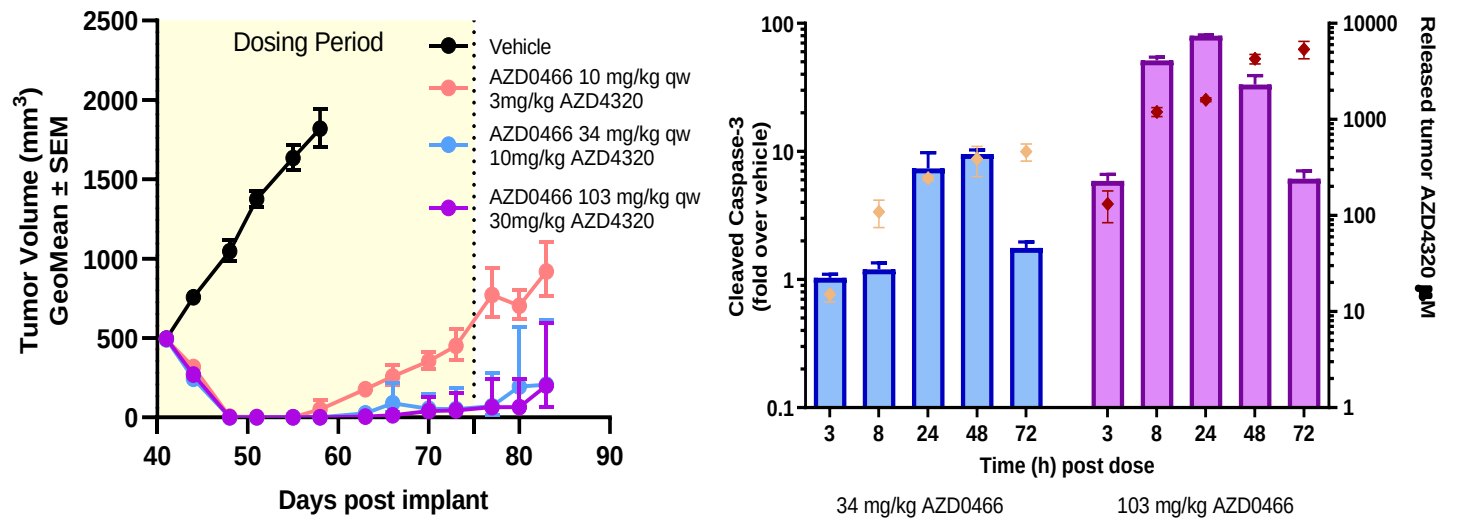
Dosing Period
Vehicle
AZD0466 10 mg/kg qw
3mg/kg AZD4320
AZD0466 34 mg/kg qw
10mg/kg AZD4320
AZD0466 103 mg/kg qw
30mg/kg AZD4320
Tumor Volume (mm3)
GeoMean ± SEM
Days post implant
Cleaved Caspase-3
(fold over vehicle)
Released tumor AZD4320 nM
Time (h) post dose
34 mg/kg AZD0466
103 mg/kg AZD0466

B: AML PDX xenograft

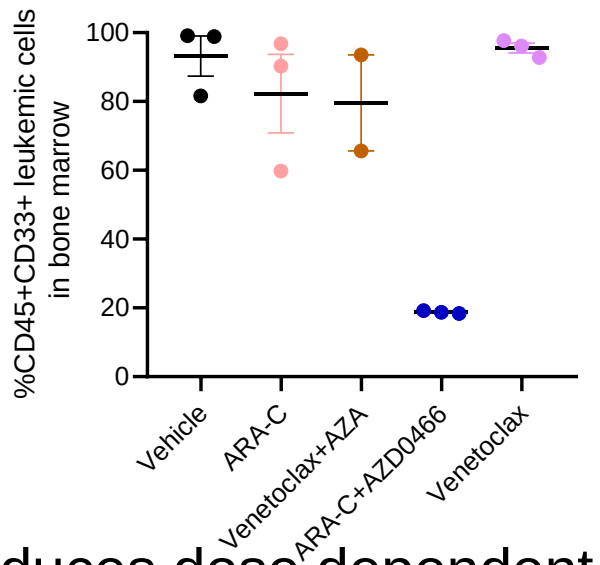
%CD45+CD33+ leukemic cells
in bone marrow
Vehicle
ARA-C
Venetoclax+AZA
ARA-C+AZD0466
Venetoclax

(A) AZD0466 induces dose dependent tumor regression and cleaved caspase 3 induction. (B) AZD0466 in combination with AraC decreases tumor burden in bone marrow of mice implanted with primary AML cells.

AZD0466 is more efficacious in SCLC PDX models compared to standard-of-care

A: SCLC PDX screen

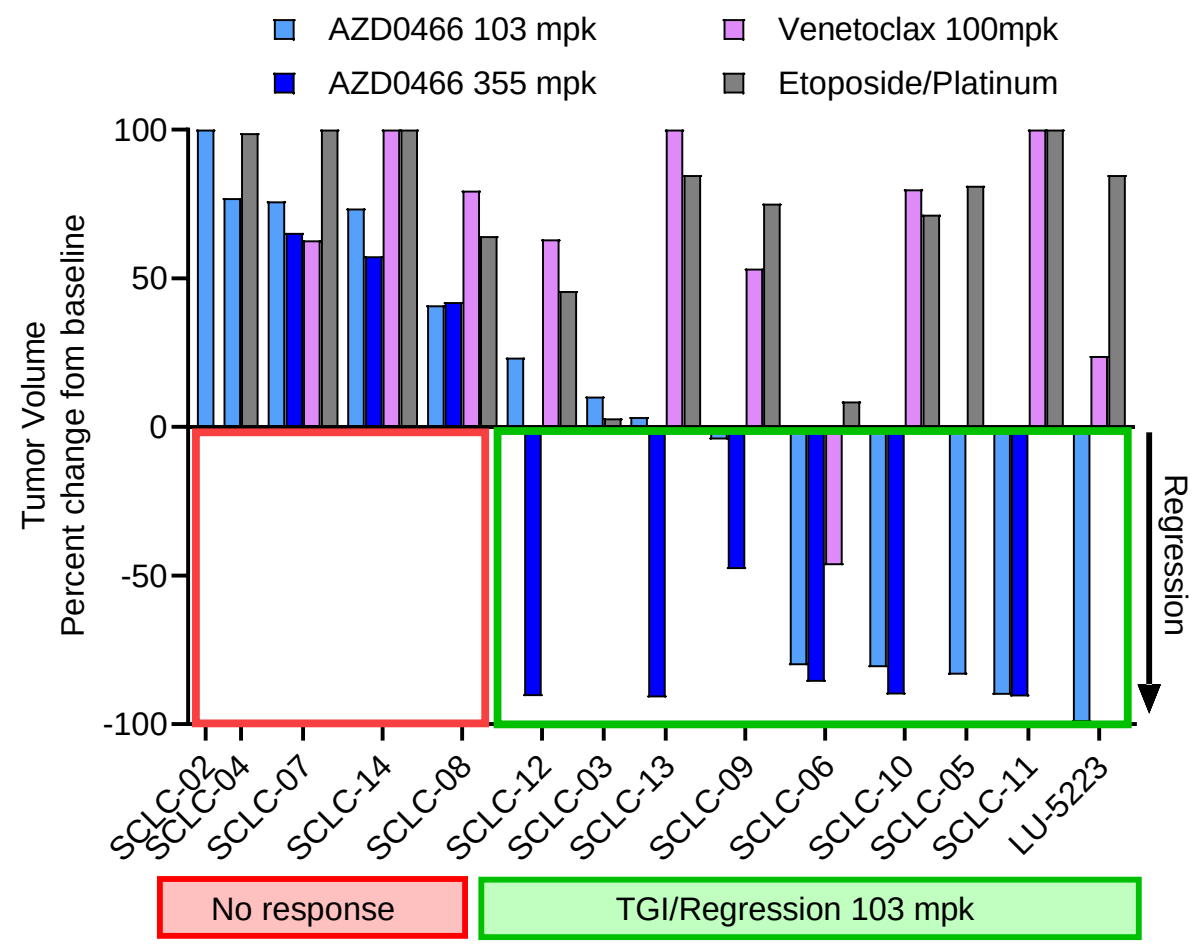
AZD0466 103 mpk
AZD0466 355 mpk
Venetoclax 100mpk
Etoposide/Platinum
Tumor Volume
Percent change fom baseline
Regression
SCLC-02
SCLC-04
SCLC-07
SCLC-14
SCLC-08
SCLC-12
SCLC-03
SCLC-13
SCLC-09
SCLC-06
SCLC-10
SCLC-05
SCLC-11
LU-5223
No response
TGI/Regression 103 mpk

B: SCLC molecular analysis

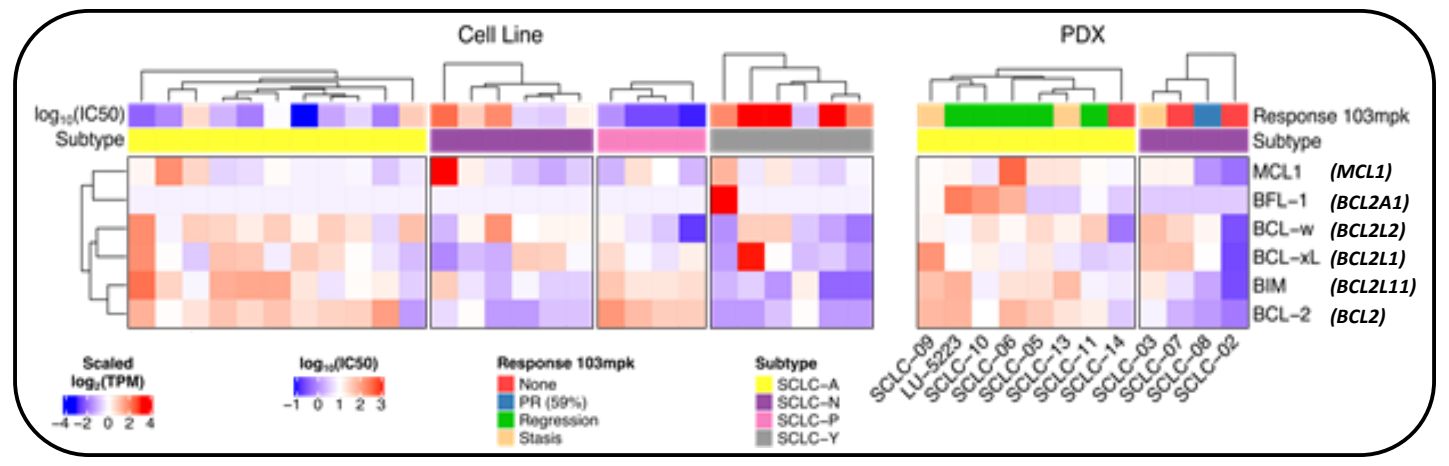
Cell Line
PDX
log10(IC50)
Subtype
Response 103mpk
MCL1 (MCL1)
BFL-1 (BCL2A1)
BCL-w (BCL2L2)
BCL-xL (BCL2L1)
BIM (BCL2L11)
BCL-2 (BCL2)
Scaled
log2(TPM)
None
PR (59%)
Regression
Stasis
SCLC-A
SCLC-N
SCLC-P
SCLC-Y

(A) Monotherapy activity of AZD0466 in a population-based PDX screen. (B) AZD0466 response is predominantly associated with SCLC-A and SCLC-P subtypes in cell lines and PDX models

AZD0466 combinations drive deep and durable response in preclinical DLBCL models

A: OCI-LY10 ABC DLBCL xenograft

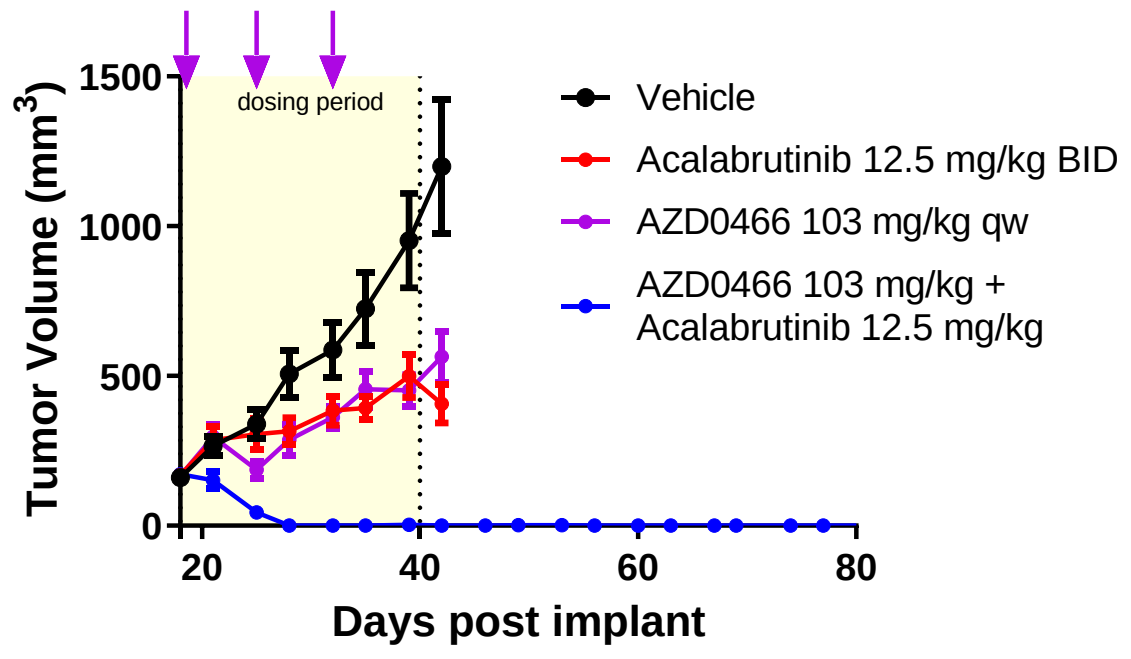
dosing period
Vehicle
Acalabrutinib 12.5 mg/kg BID
AZD0466 103 mg/kg qw
AZD0466 103 mg/kg +
Acalabrutinib 12.5 mg/kg
Tumor Volume (mm3)
Days post implant

B: SUDHL-4 GCB DLBCL xenograft

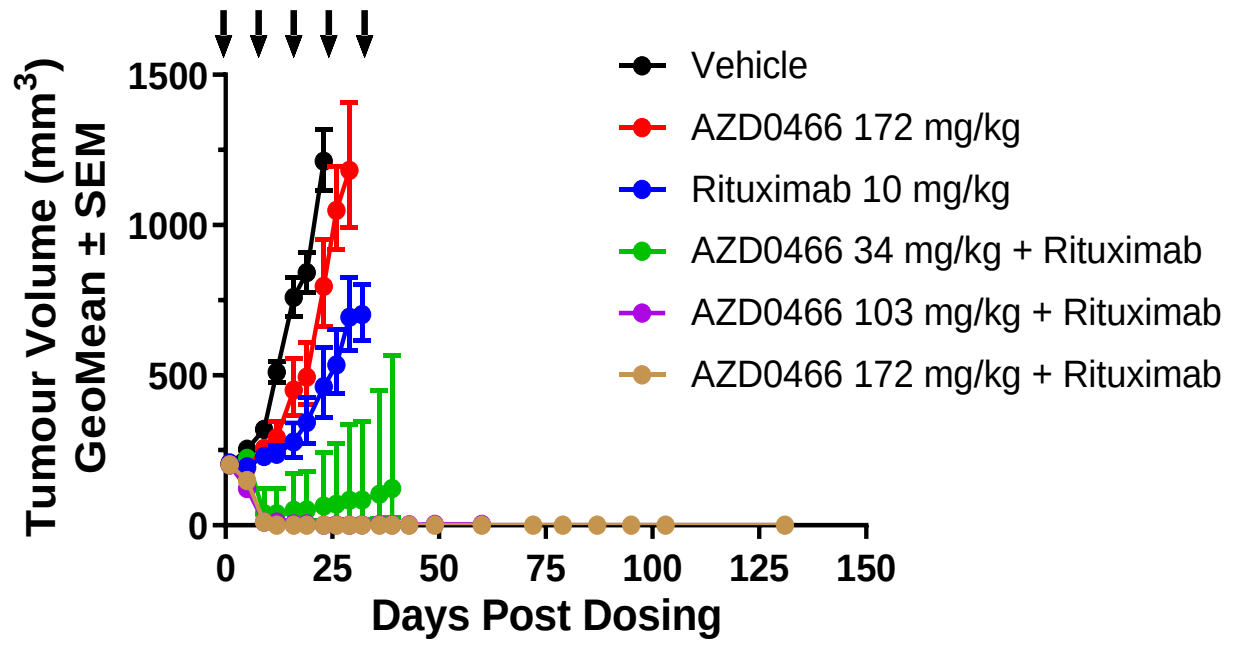
Vehicle
AZD0466 172 mg/kg
Rituximab 10 mg/kg
AZD0466 34 mg/kg + Rituximab
AZD0466 103 mg/kg + Rituximab
AZD0466 172 mg/kg + Rituximab
Tumour Volume (mm3)
GeoMean ± SEM
Days Post Dosing

(A) Combination of AZD0466 with BTK inhibitor Acalabrutinib drives durable regression in ABC DLBCL. (B) AZD0466 combined with rituximab results in complete and durable regressions in GCB DLBCL

Conclusions

- AZD0466 and its active moiety AZD4320 demonstrate differentiated activity from the Bcl-2 selective inhibitor venetoclax in preclinical models of SCLC and AML.
- AZD0466 has potential as a combinatorial agent to increase the depth and duration of response to novel and standard of care therapies, as demonstrated in models of DLBCL.

References

1. Design and optimization of a dendrimer-conjugated dual Bcl-2/Bcl-xL inhibitor, AZD0466, with improved therapeutic index, Ashford M et al, AACR (2020)

Presented at the AACR Virtual Annual Meeting II:E-Poster Submission BCL-2 Family and Mitochondrial Apoptosis Session P-56, 22-23 June 2020

Supported by AstraZeneca

REMOVE IF NOT PERMITTED